Safety and Immunogenicity of Quadrivalent Meningococcal Polysaccharide Vaccine (MPV ACYW135) Compared with Quadrivalent Meningococcal Conjugate Vaccine (Menactra®) in Malian Children

Sow, S. O.[a], Tapia, M. D.[b], Haidara, F. C.[a], Diallo, F.[a], Traore, Y.[a], Traoré, A.[a], Kodio, M.[a], Borrow, R.[c], Townsend-Payne, K.[c], Yuan, L.[d], Yang, S.[d], Shi, L.[d], Chen, J.[d], Fang, G.[d], Lin, J.[d], Hu, R.[d], Viviani, S.[e], & Huang, Z.[d]

a. Centre pour le Développement des Vaccins du Mali (CVD-Mali), Bamako, Mali
b. Department of Pediatrics, Division of Pediatric Infectious Diseases, Center for Vaccine Development and Global Health, University of Maryland School of Medicine, Baltimore, MD, USA
c. Vaccine Evaluation Unit, UK Health Security Agency, Public Health Laboratory, Manchester, UK
d. Walvax Biotechnology Co., Ltd., Kunming, Yunnan, China
e. Department of Molecular and Developmental Medicine, University of Siena, Siena, Italy

Introduction

Neisseria meningitidis causes significant morbidity and mortality in children and young adults worldwide. Among 12 meningococcal serogroups identified so far, the majority of meningococcal disease is caused by six serogroups namely A, B, C, X, Y and W. [1]
As recommended by WHO, the availability of significant quantities of vaccines will allow a constant supply of meningococcal vaccine containing *N. meningitidis* C and W polysaccharides for emergency stockpile to be used in reactive vaccination campaigns to control large outbreaks in African meningitis belt countries, or whenever needed. [2,3,4]

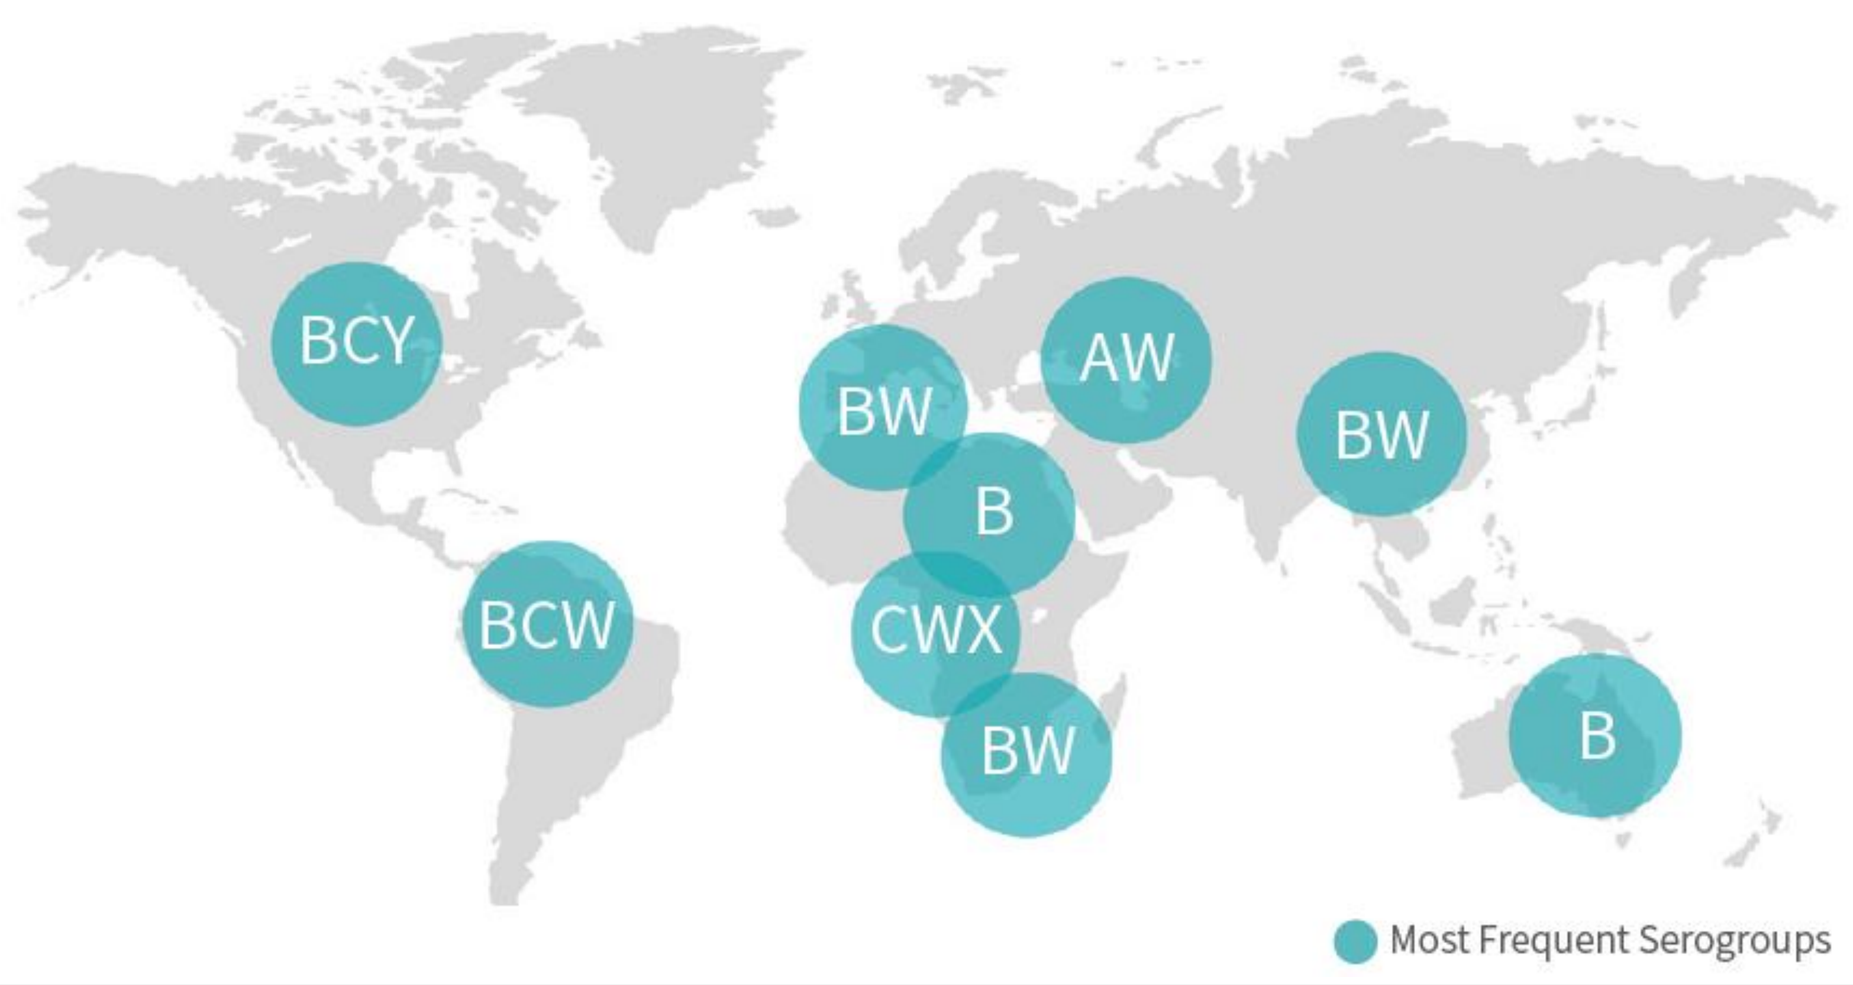
BCY
BW
AW
BW
B
BCW
CWX
BW
B
Most Frequent Serogroups

WHO: Serogroup Distribution of Invasive Meningococcal Diseases [5]

Methods

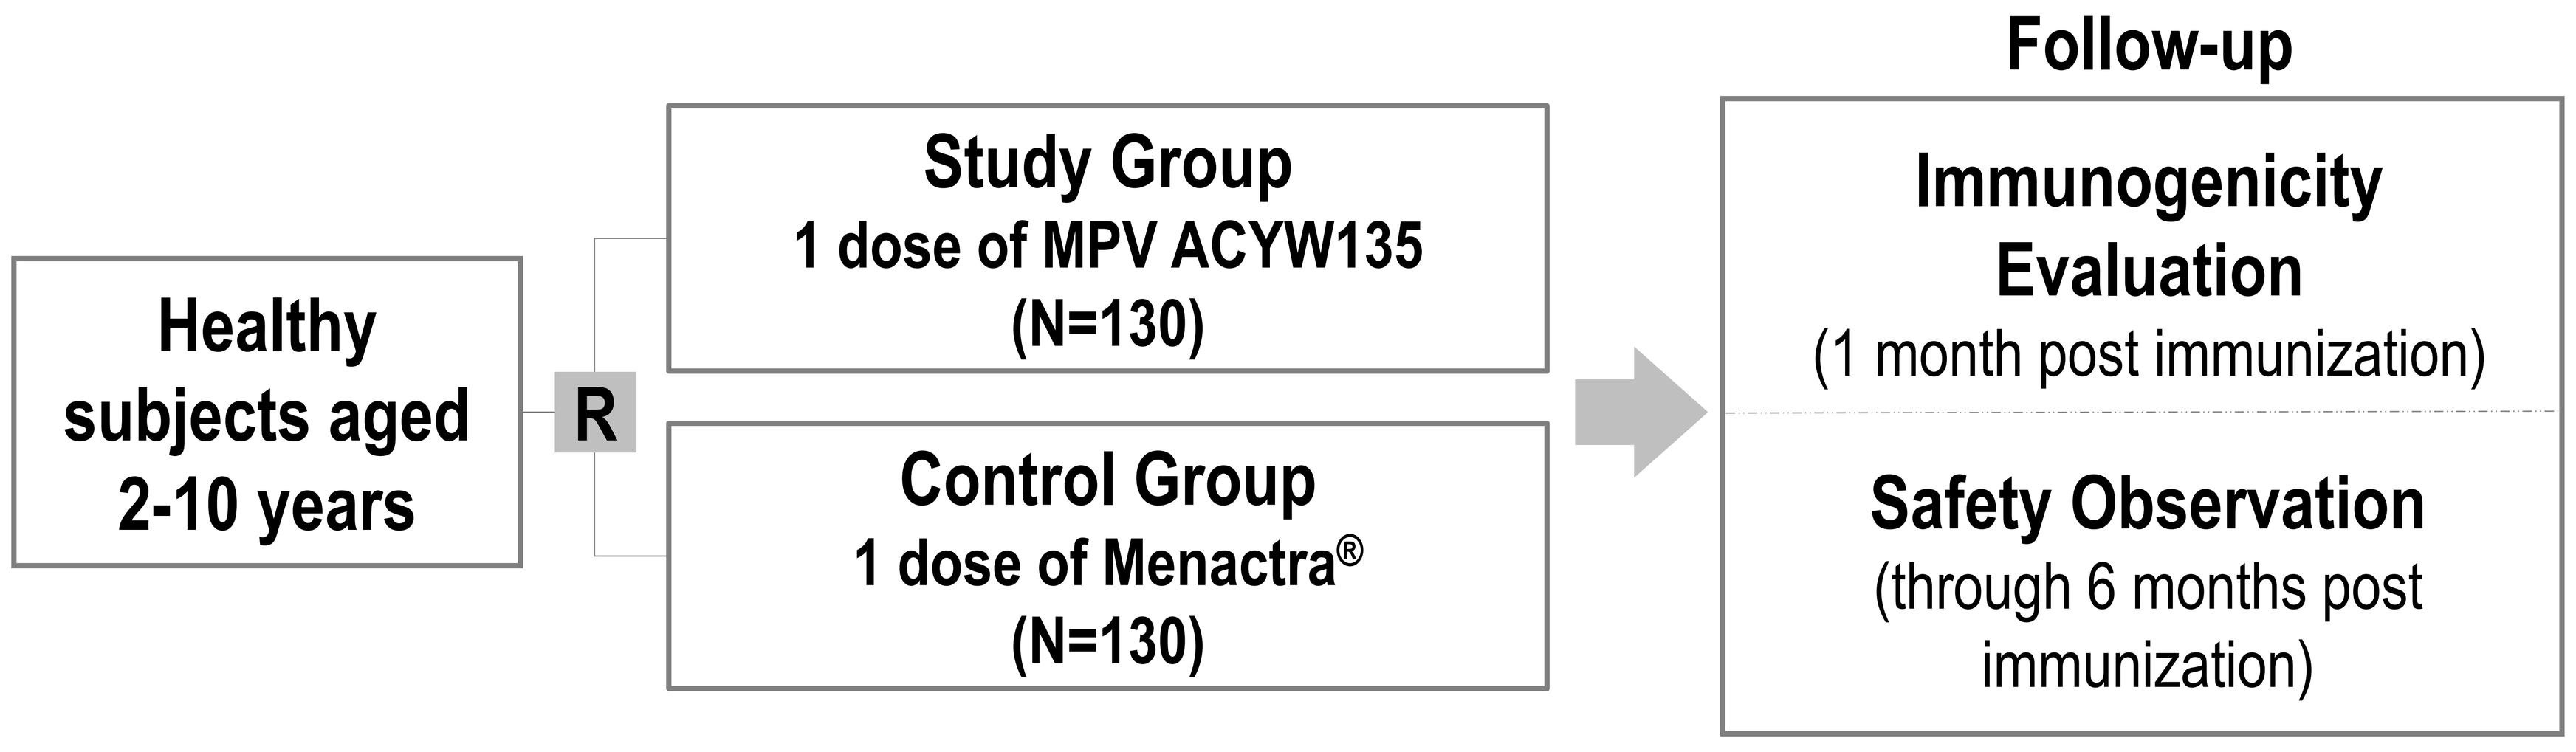
Follow-up
Healthy subjects aged 2-10 years
R
Study Group
1 dose of MPV ACYW135
(N=130)
Control Group
1 dose of Menactra®
(N=130)
Immunogenicity Evaluation
(1 month post immunization)
Safety Observation
(through 6 months post immunization)

In this study (NCT04450498), healthy, 2- to 10-year-old children in Bamako, Mali, were randomized 1:1 to receive one dose of MPV ACYW135 or Menactra®. Safety outcomes were evaluated for 6 months post-immunization. Immunogenicity for all serogroups was assessed for noninferiority between MPV ACYW135 and Menactra® 30 days post immunization by serum bactericidal antibody assay using baby rabbit complement (rSBA) with blood samples taken on the day of vaccination and 30 days after vaccination.

Primary immunogenicity endpoint:

- Seroconversion rates as defined by the percentages of subjects with rSBA titers ≥ 128 to each of the four serogroups A, C, Y, W of *N. meningitidis* at Day 30 after vaccination along with its two-sided 95% CI

Secondary immunogenicity endpoints:

- % of subjects with a rSBA ≥ 4-fold increase 30 days after vaccination as compared to baseline unadjusted titers for all serogroups
- % of subjects with rSBA titer ≥ 8 at 30 days after vaccination
- rSBA geometric mean titers (GMTs) of specific antibodies to groups A, C, Y, and W *meningococci* in both vaccine groups

Safety endpoints:

- % of subjects with local and systemic reactions within 7 days
- % of subjects with reported AEs within 30 days
- % of subjects with reported SAEs within 6 months

Results

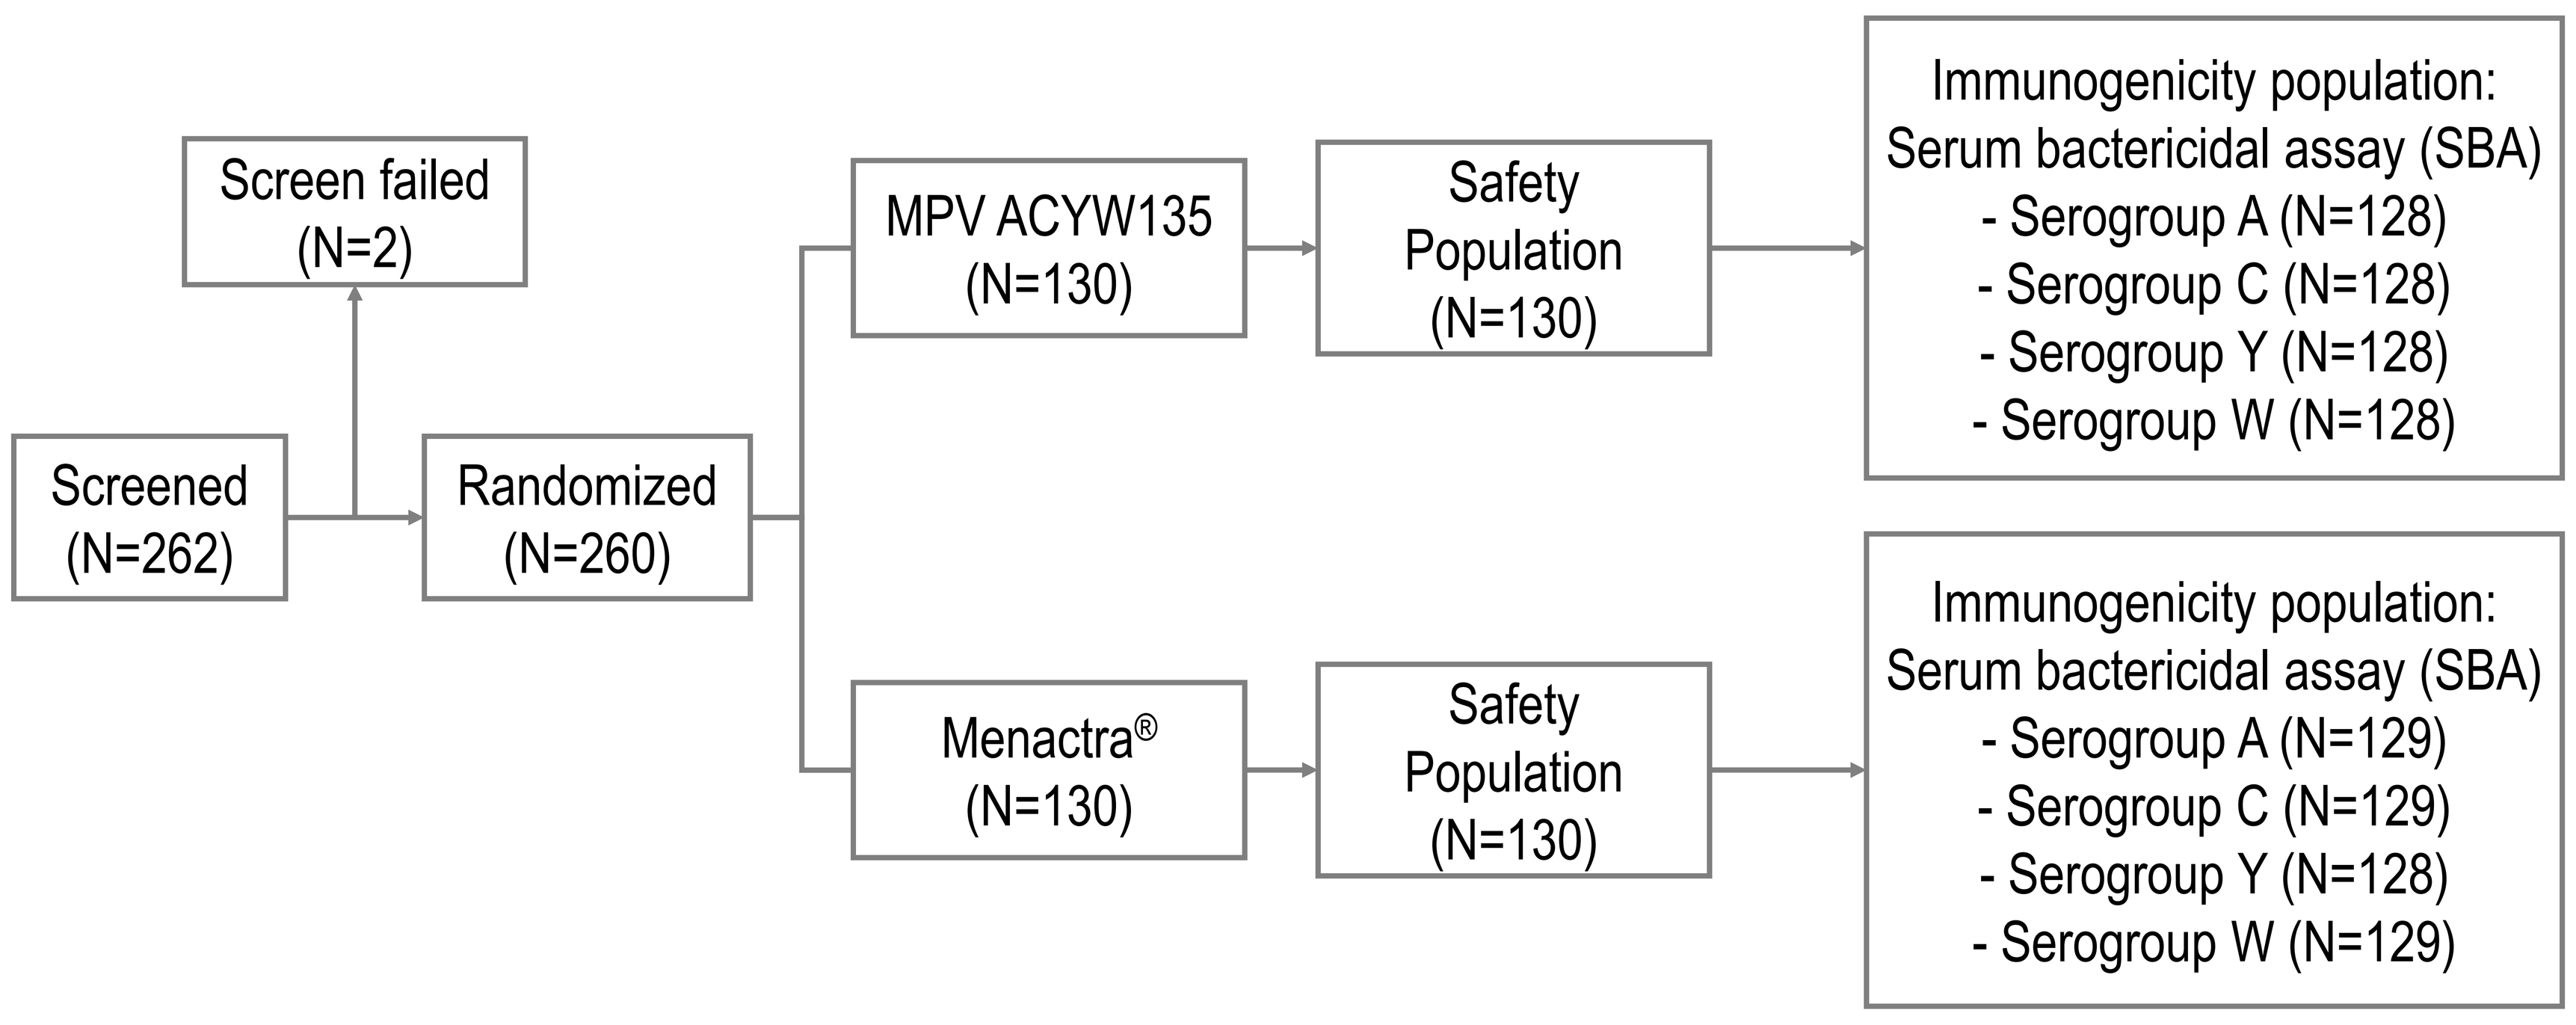
Screen failed
(N=2)
Screened
(N=262)
Randomized
(N=260)
MPV ACYW135
(N=130)
Safety Population
(N=130)
Immunogenicity population:
Serum bactericidal assay (SBA)
- Serogroup A (N=128)
- Serogroup C (N=128)
- Serogroup Y (N=128)
- Serogroup W (N=128)
Menactra®
(N=130)
Safety Population
(N=130)
Immunogenicity population:
Serum bactericidal assay (SBA)
- Serogroup A (N=129)
- Serogroup C (N=129)
- Serogroup Y (N=128)
- Serogroup W (N=129)

One dose of MPV ACYW induces non-inferior immune responses as compared to Menactra® at Day 30 post vaccination

- **Non-inferior immunogenicity** of MPV ACYW135 as compared to Menactra® was both demonstrated when comparing the percentages of subjects in each vaccine group with rSBA titers ≥ 128 (primary endpoint) and those achieving a rSBA ≥ 4-fold increase after vaccination as compared to baseline unadjusted titers (secondary endpoint)
- Proportions of subjects with rSBA titers ≥ 8 for all serogroups were similar between vaccine groups (p>0.05)
- Geometric Mean Titers (GMTs) and Geometric Mean Fold Increases (GMFIs) for all serogroups in both vaccine groups were similar between vaccine groups (p>0.05)

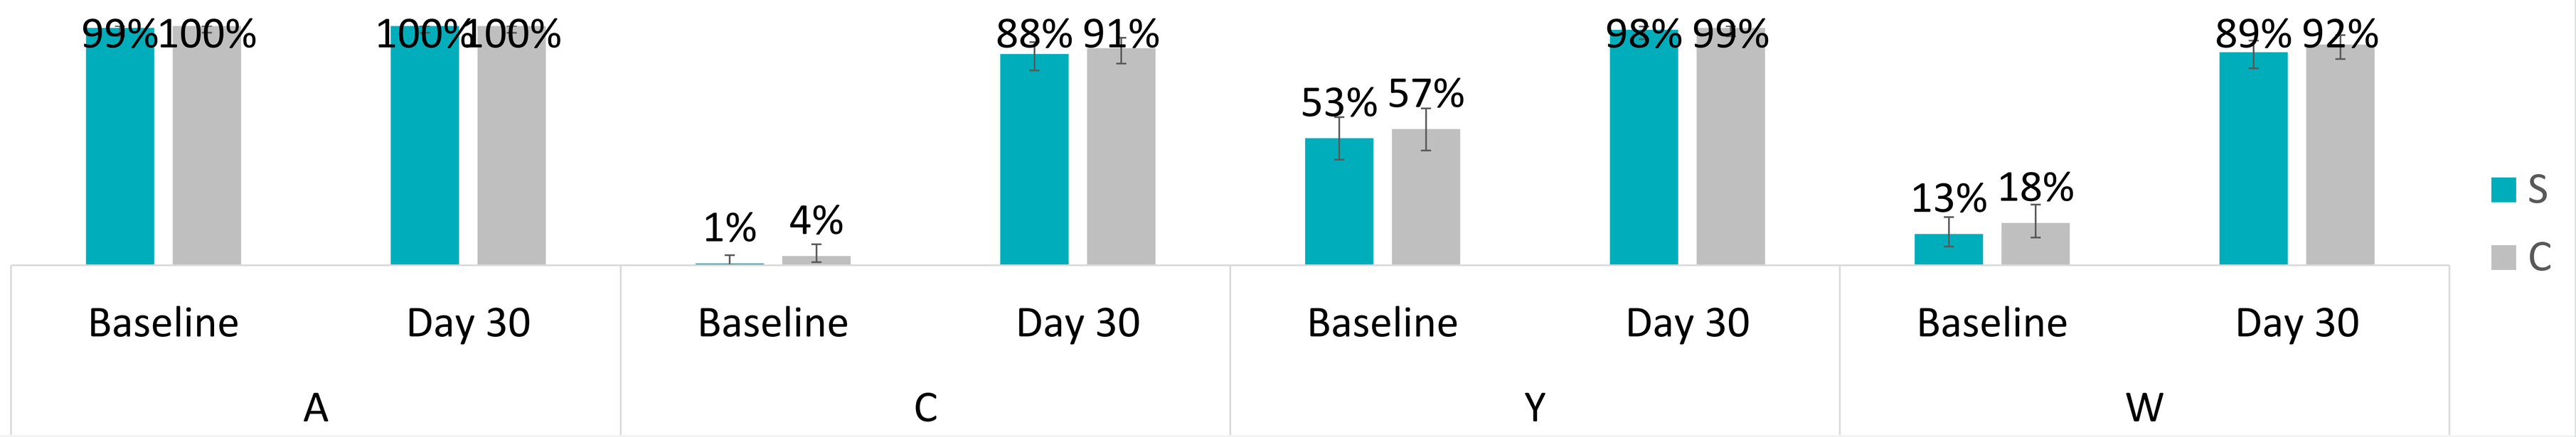
99%
100%
100%
100%
1%
4%
88%
91%
53%
57%
98%
99%
13%
18%
89%
92%
Baseline
Day 30
Baseline
Day 30
Baseline
Day 30
Baseline
Day 30
A
C
Y
W
S
C

Subjects (%) with rSBA Titers ≥ 128 at Baseline and Day 30 post Vaccination (S: Study Group; C: Control Group)

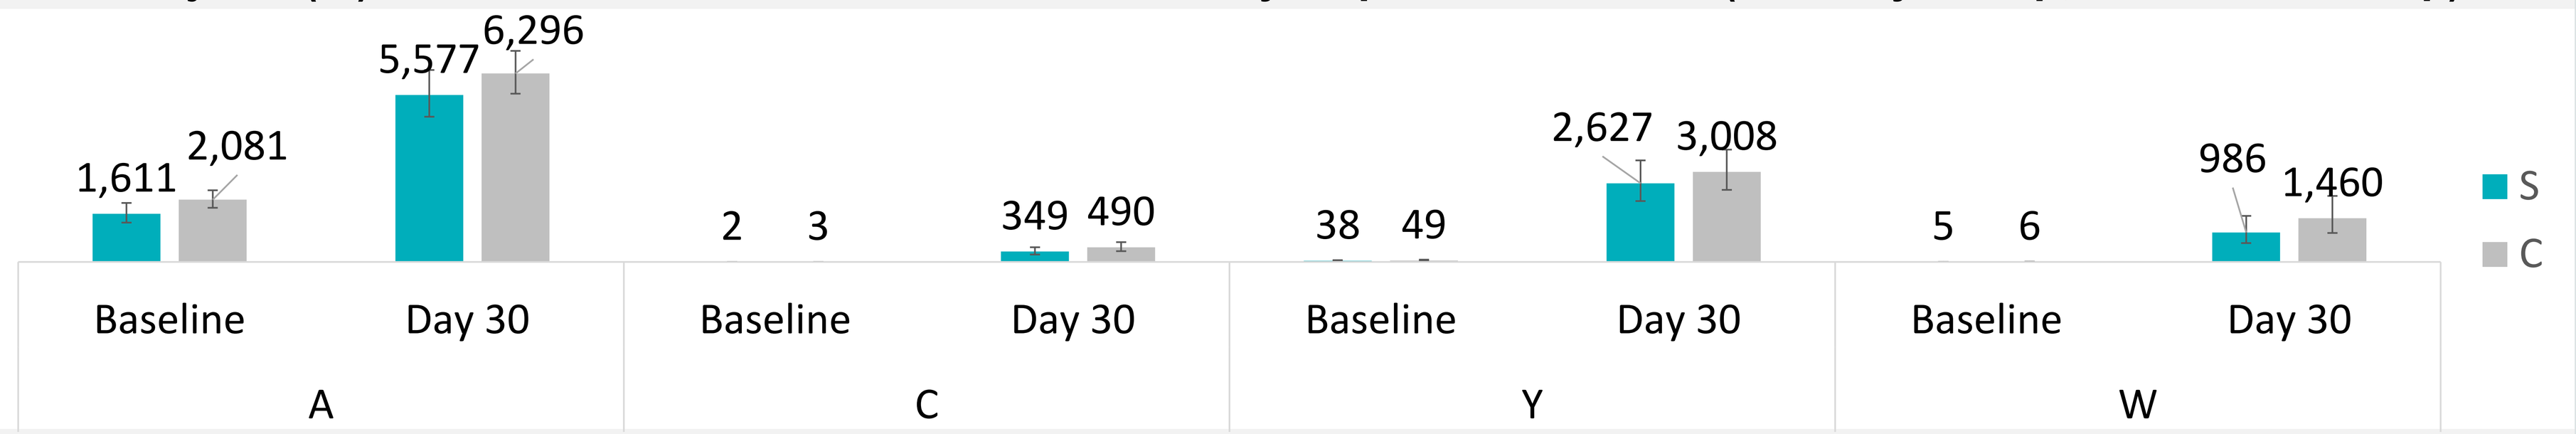
1,611
2,081
5,577
6,296
2
3
349
490
38
49
2,627
3,008
5
6
986
1,460
Baseline
Day 30
Baseline
Day 30
Baseline
Day 30
Baseline
Day 30
A
C
Y
W
S
C

rSBA Antibody GMT at Baseline and Day 30 post Immunization (S: Study Group; C: Control Group)

MPV ACYW135 showed a similar reactogenicity and safety profile to Menactra®

- No immediate post-immunization reactions were observed
- Local and systemic post-immunization reactions for 7 days after immunization were overall few and similar between vaccine groups for severity and duration (p>0.05), all of which resolved without sequelae
- Unsolicited adverse events were similar in both vaccine groups for relationship to study vaccines, severity and duration (p>0.05)
- No SAE was reported during the study period

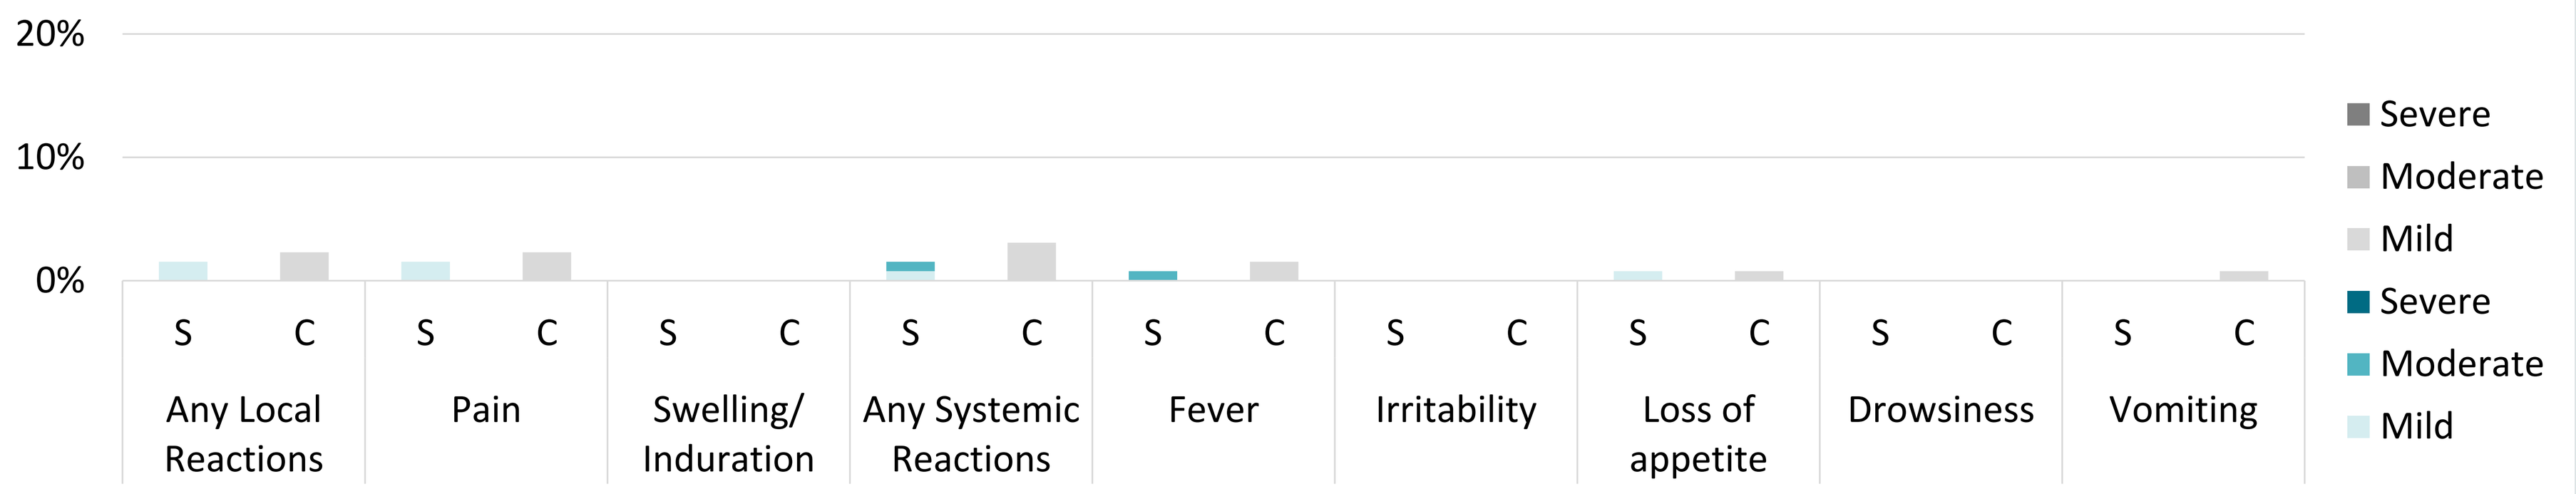
20%
10%
0%
S
C
Any Local Reactions
Pain
Swelling/ Induration
Any Systemic Reactions
Fever
Irritability
Loss of appetite
Drowsiness
Vomiting
Severe
Moderate
Mild
Severe
Moderate
Mild

Subjects (%) With Solicited Local or Systemic ARs within 30 Minutes to Day 7 post Vaccination by Severity (S: Study Group; C: Control Group)

Conclusions

This study demonstrates that MPV ACYW135 vaccine has a similar reactogenicity and immunogenicity profile to Menactra® vaccine. Therefore, MPV ACYW135 vaccine can be stockpiled and used in reactive immunization campaigns to control local outbreaks of N. meningitidis serogroups C and W in Africa and in other settings until affordable conjugate polyvalent vaccines become available.

Acknowledgments

The authors gratefully acknowledge Pharmalys Ltd. for coordinating the study in Mali; the Center of Excellence for Biomedical and Public Health Informatics (BIOPHICS) for performing data management and statistical analysis; and the Vaccine Evaluation Unit, National Infection Service, UK Health Security Agency, Manchester, UK for rSBA testing, respectively. Finally, the authors express their gratitude to the children who took part in this study along with their parents, for their support and commitment.

Funding Source

This work was supported by Yuxi Walvax Biotechnology Co., Ltd., Yuxi, Yunnan, China.

References:
1. Identification of N. meningitidis serogroup and performing the SASG test 2021. 2021 [accessed 2021 Jan 27]. https://dermatologyadvisor.blogspot.com/2019/01/identification-of-n-meningitidis.html
2. WHO. Epidemic meningitis control in countries of the African meningitis belt, 2016. WHO Wkly Epidemiol Rec. 2017 Nov 13;92:145–64.8.
3. WHO. Continuing risk of meningitis due to Neisseria meningitidis serogroup C in Africa: revised recommendations from a WHO expert consultation. WHO Wkly Epidemiol Rec. 2017 Nov 41;92:609–24.
4. WHO. Meningococcal vaccines: WHO position paper, November 2011. Wkly Epidemiol Rec. 2011 Nov 18;86(47):521–39
5. WHO (2019). Invasive Meningococcal Disease-Serotype Distribution. Available at: https://www.who.int/health-topics/meningitis#tab=tab_1

WALVAX BIOTECHNOLOGY CO., LTD.